onset 1: efficacy and safety of mealtime fast-acting insulin aspart versus insulin aspart after 52 weeks

1. **Chantal Mathieu**
 UZ Leuven, Leuven, Belgium
2. **Bruce Bode**
 Atlanta Diabetes Associates, Atlanta, Georgia, USA
3. **Edward Franek**
 Mossakowski Medical Research Center, Polish Academy of Sciences, Warsaw, Poland
4. **Athena Philis-Tsimikas**
 Scripps Whittier Diabetes Institute, San Diego, California, USA
5. **Ludger Rose**
 Diabetes Research Center, Münster, Germany
6. **Tina Graungaard**
 Novo Nordisk A/S, Søborg, Denmark
7. **Anne Birk Østerskov**
 Novo Nordisk A/S, Søborg, Denmark
8. **Shazia Azizuddin**
 Novo Nordisk Ltd., Gatwick, UK
9. **David Russell-Jones**
 Royal Surrey County Hospital, Guildford, UK

Introduction

- onset 1 was a phase 3a, randomised, partially double-blind trial evaluating the efficacy and safety of fast-acting insulin aspart (faster aspart) in adults with type 1 diabetes (T1D) over 52 weeks, in two 26-week treatment periods (Russell-Jones *et al. Diabetes Care* 2017;40:943–50).
- At 26 weeks, non-inferiority to insulin aspart (IAsp) for HbA_{1c} change from baseline was confirmed for both mealtime and post-meal faster aspart (Russell-Jones *et al. Diabetes Care* 2017;40:943–50).
 - A statistically significant benefit for 2-h postprandial glucose (PPG) increment (meal test) was also confirmed for mealtime faster aspart versus mealtime IAsp.
 - No statistically significant difference was reported in overall rate of severe or blood glucose (BG)-confirmed hypoglycaemia.

Aim

- To assess the long-term safety and efficacy of mealtime faster aspart over 52 weeks.

Methods

- In the initial 26-week treatment period of onset 1, subjects with T1D were randomised to either double-blind mealtime faster aspart, IAsp or open-label post-meal faster aspart, each with insulin detemir. Subjects on mealtime faster aspart (n=381) and mealtime IAsp (n=380) continued to the additional 26-week treatment period.

Statistical analysis

- For efficacy endpoints, change from baseline was analysed using a mixed-effect model for repeated measurements, including treatment, region and strata as fixed effects, subject as random effect, baseline as covariate, and interaction between fixed effects and visit, and between the covariate and visit.
- Severe or BG-confirmed hypoglycaemic episodes were analysed using a negative binomial model with log-link function and the logarithm of the time in which the hypoglycaemic episode is considered treatment-emergent as offset. The model includes treatment, region and strata as factors.

Results

Baseline characteristics

- Baseline characteristics were similar in both groups. The mean age of the sample was 46.1 years, with a mean HbA_{1c} at baseline of 7.62% (range: 6.0–9.8%) and 7.58% (range: 5.6–9.6%) in the mealtime faster aspart and IAsp groups, respectively. The mean durations of diabetes were 20.9 years and 19.3 years in the mealtime faster aspart and IAsp groups, respectively.

Efficacy

- After 52 weeks, HbA_{1c} change from baseline was statistically significantly greater for faster aspart versus IAsp (–0.08% vs. +0.01%, respectively; estimated treatment difference [ETD] [95% confidence interval {CI}]: –0.10% [–0.19;–0.00]; *p*=0.0424) (Figure 1).
- Estimated change from baseline in 1-h PPG increment (meal test) was significant in favour of faster aspart: –1.05 mmol/L vs. –0.14 mmol/L with IAsp (ETD: –0.91 mmol/L [–1.40; –0.43]; *p*=0.0002) (Figure 3).
 - There were no statistically significant treatment differences in 2-, 3- and 4-h PPG increments.

Figure 1 HbA_{1c} over time.

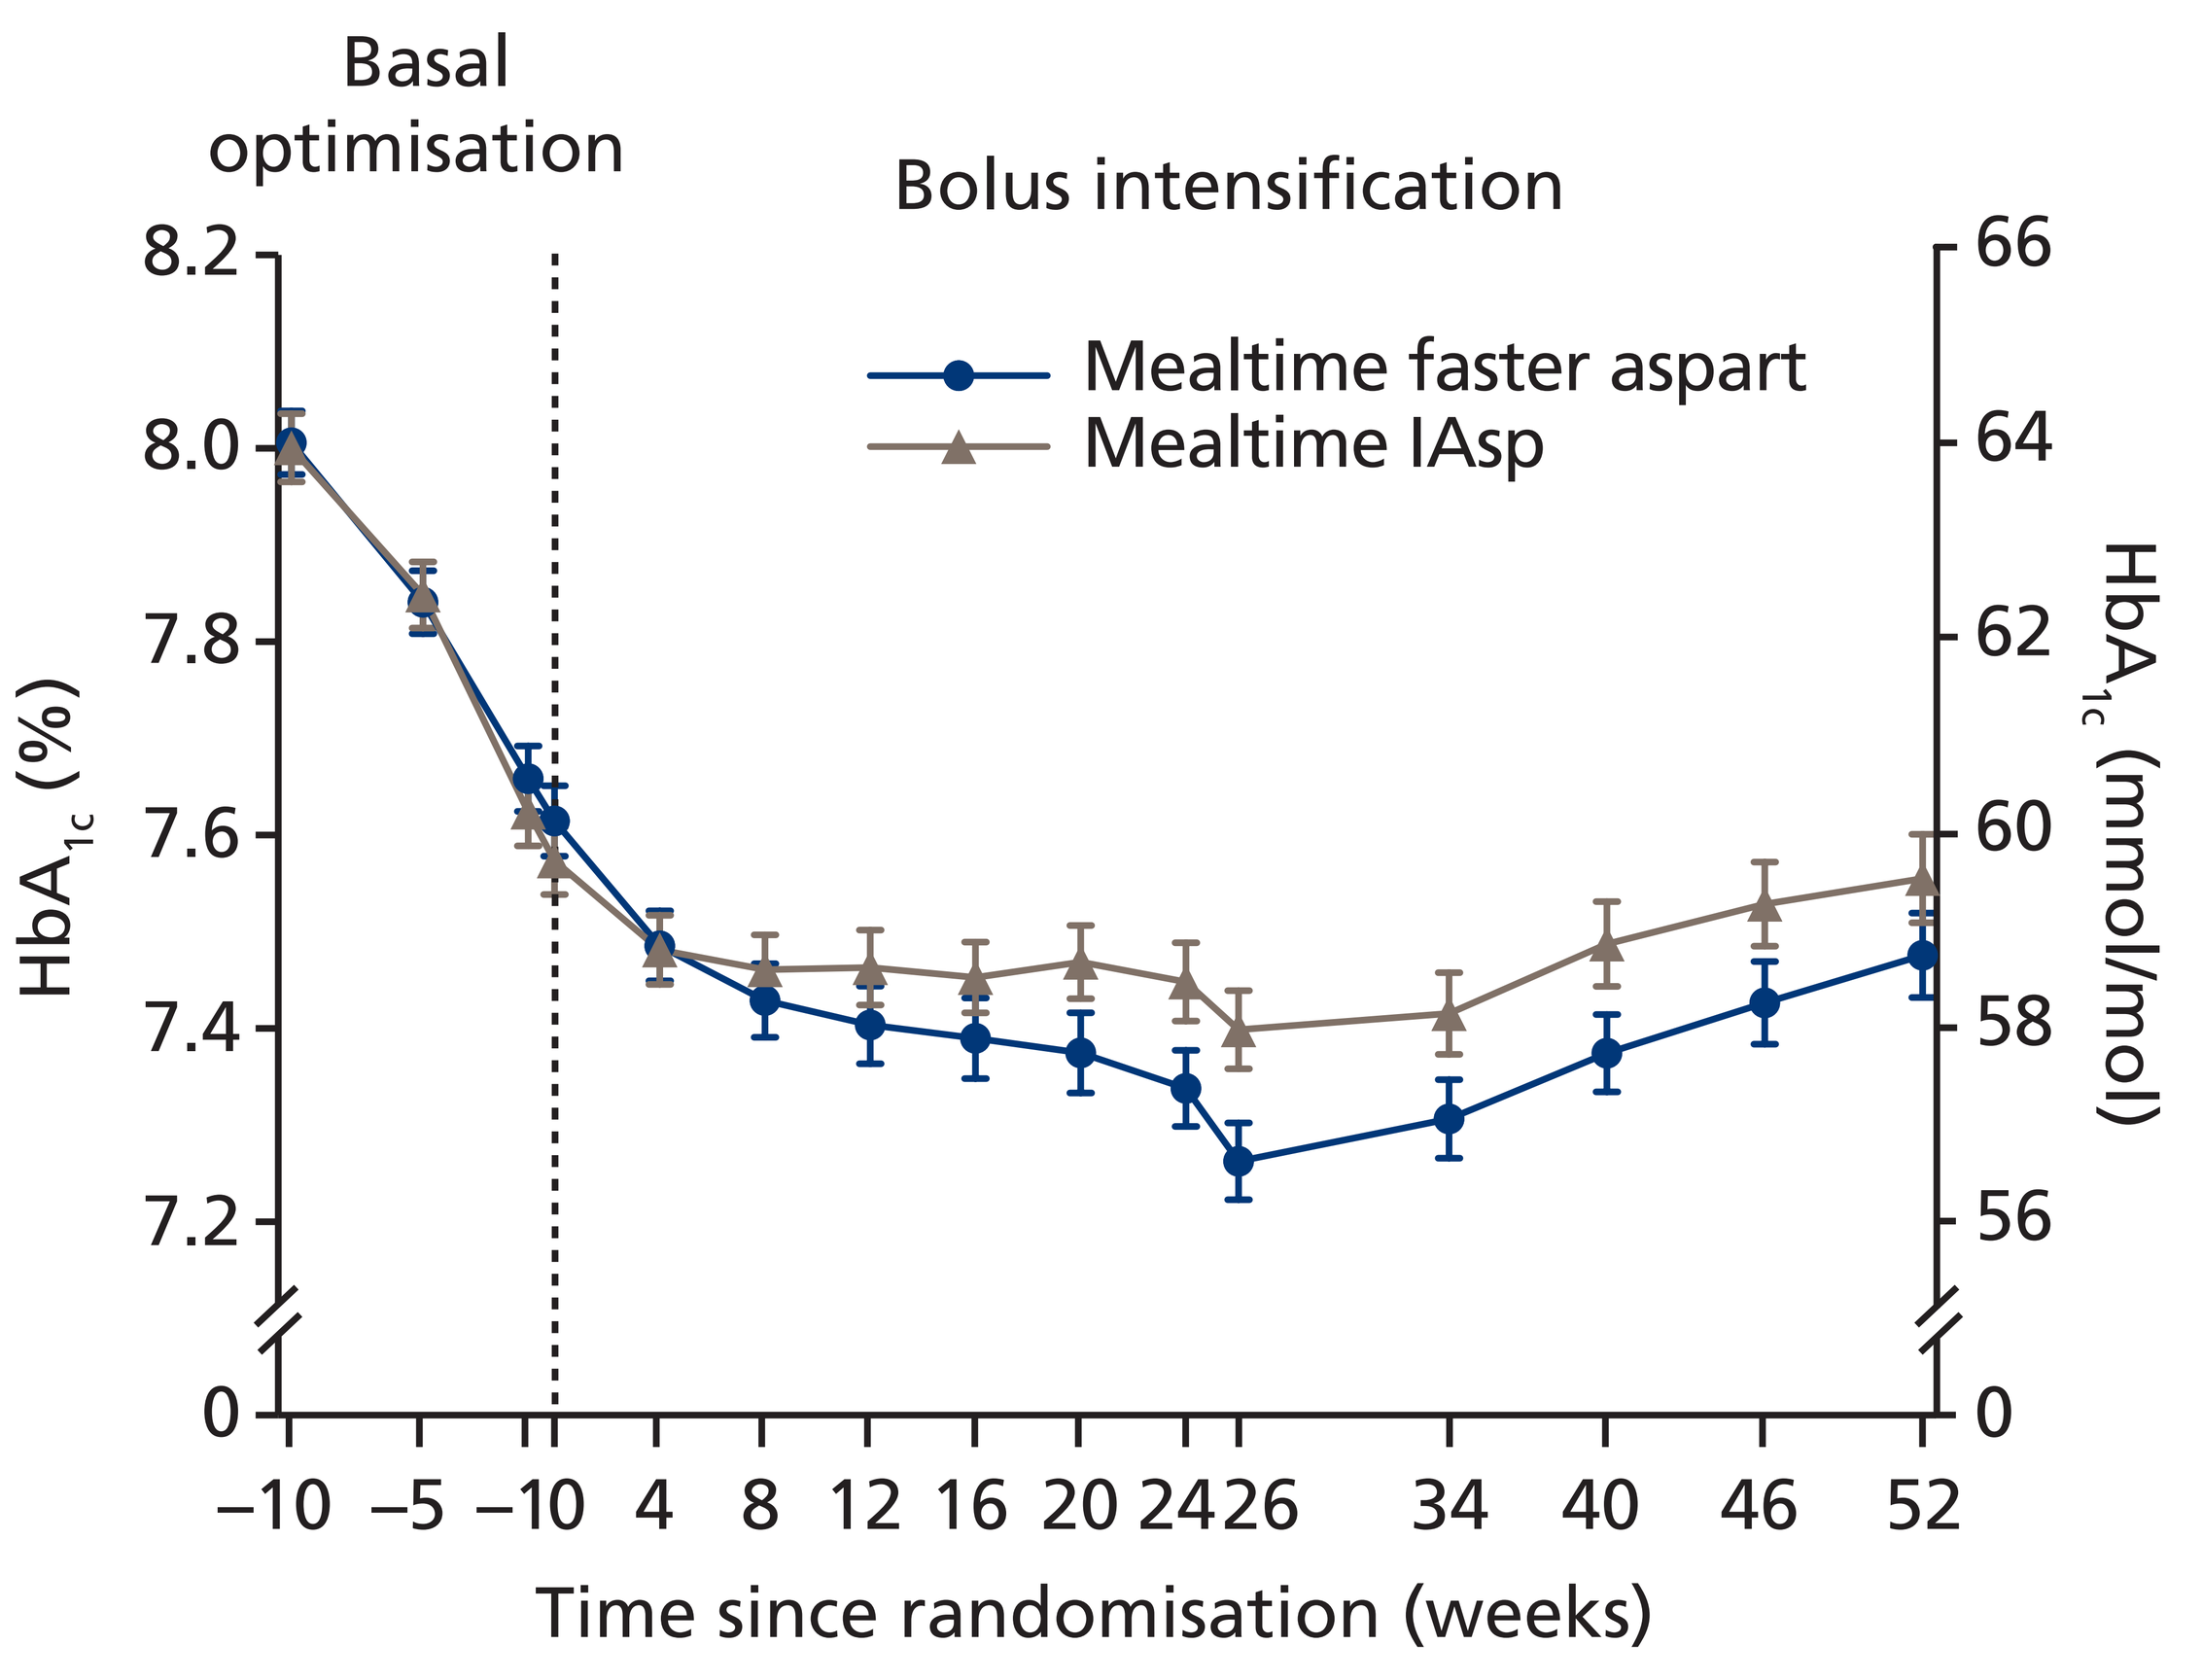

Basal optimisation
Bolus intensification
Mealtime faster aspart
Mealtime IAsp
HbA1c (%)
HbA1c (mmol/mol)
8.2
8.0
7.8
7.6
7.4
7.2
0
66
64
62
60
58
56
0
–10 –5 –10 4 8 12 16 20 24 26 34 40 46 52
Time since randomisation (weeks)

Error bars ± standard error (mean). Faster aspart, fast-acting insulin aspart; IAsp, insulin aspart.

Figure 2 PPG increment (meal test) at week 52.

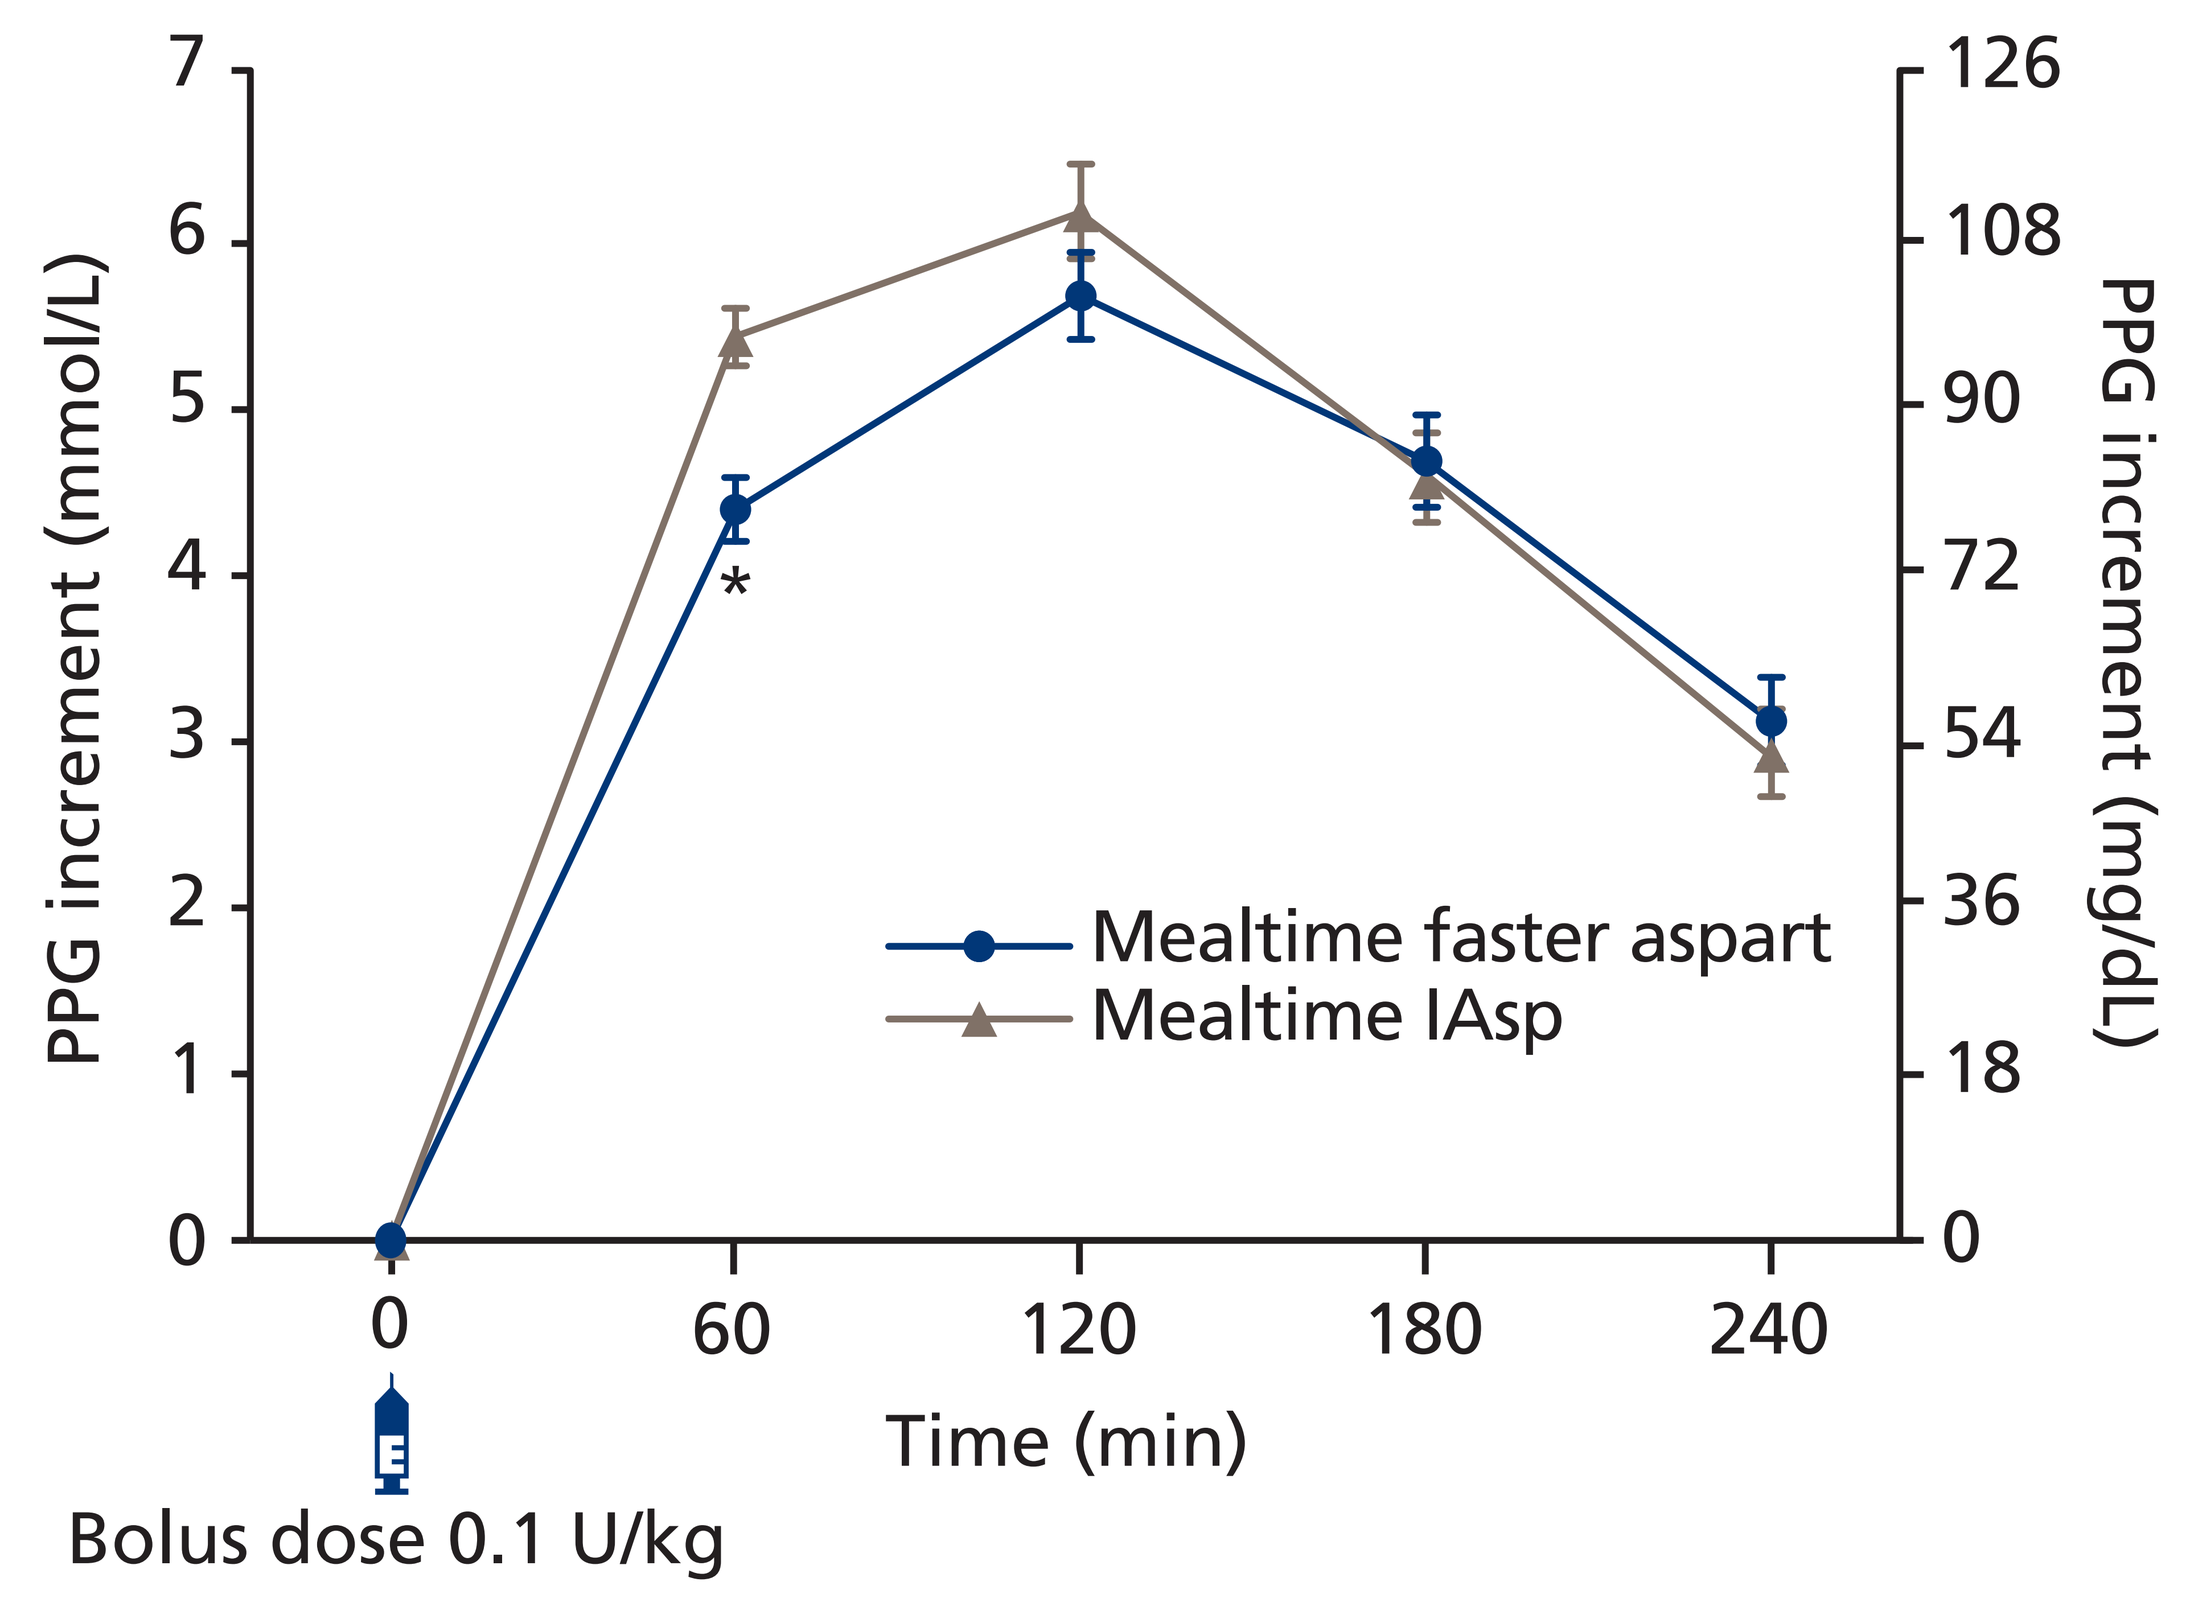

PPG increment (mmol/L)
PPG increment (mg/dL)
7
6
5
4
3
2
1
0
126
108
90
72
54
36
18
0
0 60 120 180 240
Time (min)
Bolus dose 0.1 U/kg
*
Mealtime faster aspart
Mealtime IAsp

Error bars ± standard error (mean). *p=0.0002. Faster aspart, fast-acting insulin aspart; IAsp, insulin aspart; PPG, postprandial glucose.

Figure 3 9-point SMPG profile at baseline and week 52.

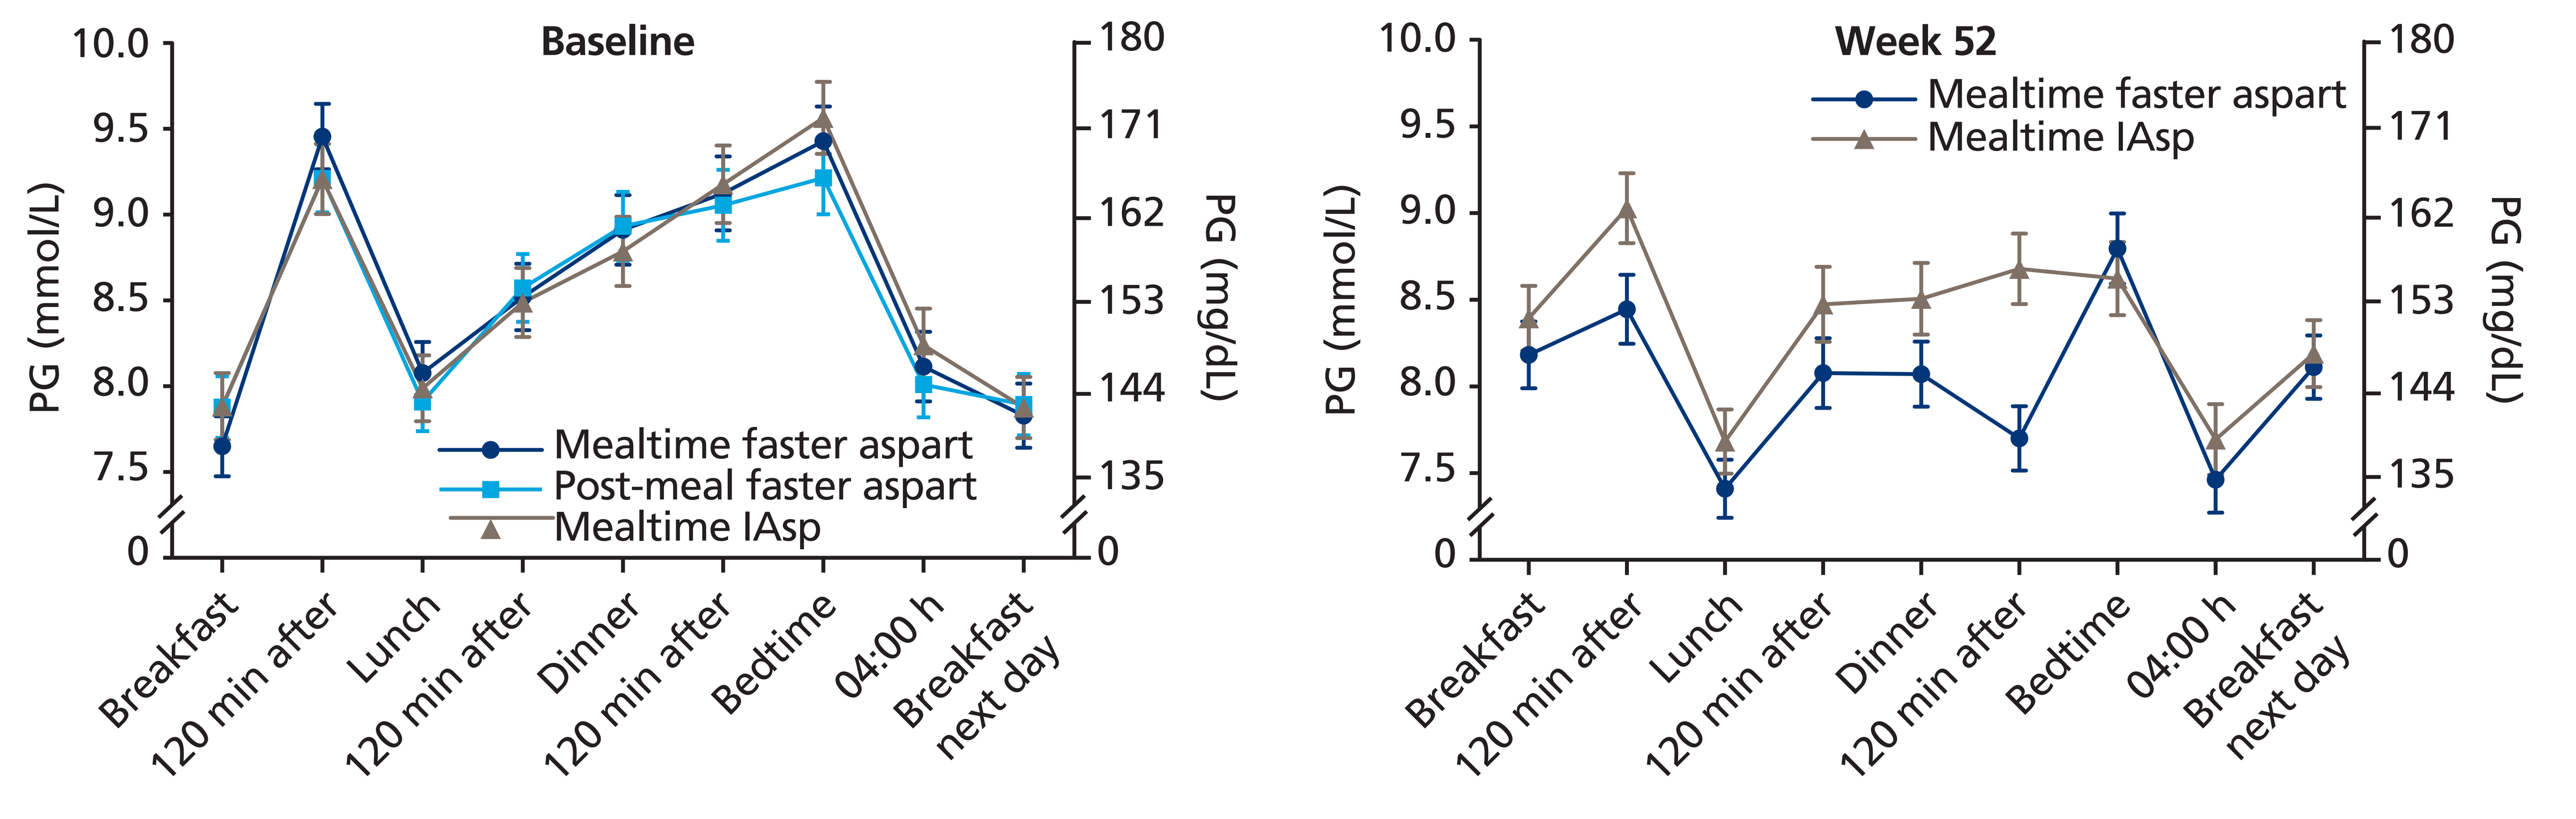

Baseline
Week 52
PG (mmol/L)
PG (mg/dL)
10.0
9.5
9.0
8.5
8.0
7.5
0
180
171
162
153
144
135
0
Mealtime faster aspart
Post-meal faster aspart
Mealtime IAsp
Breakfast
120 min after
Lunch
120 min after
Dinner
120 min after
Bedtime
04:00 h
Breakfast next day

Error bars ± standard error (mean). Faster aspart, fast-acting insulin aspart; IAsp, insulin aspart; PG, plasma glucose; SMPG, self-measured plasma glucose.

- Estimated change from baseline in mean 7-9-7-point self-measured plasma glucose (SMPG) profile was significant in favour of faster aspart: –0.41 mmol/L vs. –0.18 mmol/L with IAsp (ETD: –0.23 mmol/L [–0.46;–0.00]; *p*=0.047) (Figure 4).
 - Significant treatment differences in favour of mealtime faster aspart over IAsp for 2-h PPG, based on the 7-9-7-point SMPG profiles, were seen for breakfast (–0.62 mmol/L [–1.04;–0.20]; *p*=0.0036), main evening meal (–0.42 mmol/L [–0.80;–0.03]; *p*=0.0354) and across all meals (–0.40 mmol/L [–0.68;–0.11]; *p*=0.0067), along with 2-h PPG increments at breakfast (–0.44 mmol/L [–0.87;–0.01]; *p*=0.0462) and for main evening meal (–0.47 mmol/L [–0.89;–0.05]; *p*=0.0269).
- Estimated change from baseline in fasting plasma glucose was similar: +0.29 mmol/L for faster aspart and +0.23 mmol/L for IAsp (ETD: 0.07 mmol/L [–0.39;0.53]).
- Estimated change from baseline in 1,5-anhydroglucitol (1,5-AG) was significant in favour of faster aspart: +0.50 µg/mL vs. +0.15 µg/mL with IAsp (ETD: 0.35 µg/mL [0.05;0.65]; *p*=0.0243), reflecting less hyperglycaemic excursion with faster aspart.
- Body weight increase was similar with faster aspart (+1.18 kg) and IAsp (+1.05 kg) (ETD: 0.13 kg [–0.38;0.65]).

Safety

- The overall safety profile, including treatment-emergent adverse events (TEAEs), immunogenicity and standard safety parameters, was similar between treatments, and as expected for IAsp.
- The proportion of subjects reporting TEAEs was similar between faster aspart (83.9%) and IAsp (84.2%), with rates of 4.46 and 4.11 events per patient-year of exposure (PYE), respectively.
- There was no difference in overall severe or BG-confirmed hypoglycaemia rates between treatments (faster aspart, 53.29 per PYE, IAsp, 53.19 per PYE; estimated rate ratio: 1.01 [0.88;1.15]).
- The rate of severe hypoglycaemic episodes was 0.18 and 0.23 per PYE for faster aspart and IAsp, respectively.

Conclusions

- No long-term safety issues were identified with faster aspart.
- Overall glycaemic control remained significantly improved after 52 weeks with faster aspart versus IAsp.
- Improvement in postprandial glucose control was reflected in meal test, SMPG and 1,5-AG results.
- Approaching a profile closer to physiology with faster aspart achieves lower PPG and HbA_{1c} in T1D compared with IAsp.

The study was sponsored by Novo Nordisk and is registered with ClinicalTrials.gov (NCT01831765).
The authors acknowledge the medical writing assistance of Watermeadow Medical, an Ashfield company.
Presented at the Association of British Clinical Diabetologists Autumn Meeting, 9–10 November 2017, London, UK.